P0712 - Integrated summary of safety of obefazimod in Phase 3 ABTECT induction trials

European Crohn's and Colitis Organisation
This poster was presented at the annual ECCO Congress.
to access *all* obefazimod publications at ECCO26

Ursula Seidler[1], Silvio Danese[2], Laurent Peyrin-Biroulet[3], Marla Dubinsky[4], Raja Atreya[5], Alessandro Armuzzi[6], Fabio Cataldi[7], Doug Jacobstein[7], Christopher J Rabbat[7], Kevin Shan[7], David Laharie[8], Parambir S Dulai[9], David T Rubin[10], Bruce E Sands[11]

[1] Medizinische Hochschule Hannover, Germany, [2]Gastroenterology and Endoscopy IRCCS Ospedale San Raffaele, Italy, [3]INFINY Institute, INSERM NGERE, CHRU Nancy, Vandœuvre-lès-Nancy, France, [4]Pediatric GI and Nutrition Mount Sinai Kravis Children's Hospital, New York, USA, [5] University Hospital Erlangen, Germany, [6] IRCCS Humanitas Research Hospital in Milan, Italy, [7]Abivax, Paris, France, [8]CHU Bordeaux, Haut-Lévêque hospital, France, [9] Feinberg School of Medicine Northwestern University, Chicago, USA, [10]University of Chicago Medicine, Chicago, USA, [11] Icahn School of Medicine at Mount Sinai, New York, USA.

Background

- Obefazimod (Obe) is an oral, once-daily (QD), small molecule that enhances expression of microRNA-124, which restores mucosal immune balance through regulation of Th17 cells and macrophages. Obe has shown efficacy in patients (pts) with moderately to severely active ulcerative colitis (UC) [1-3].
- In the two Phase 3 ABTECT induction trials (Fig. 1), Obe achieved clinically meaningful improvements in clinical, endoscopic and histologic endpoints.
- Here, we report an integrated safety analysis of Obe in the two ABTECT trials.

Fig. 1: Design of ABTECT induction trials

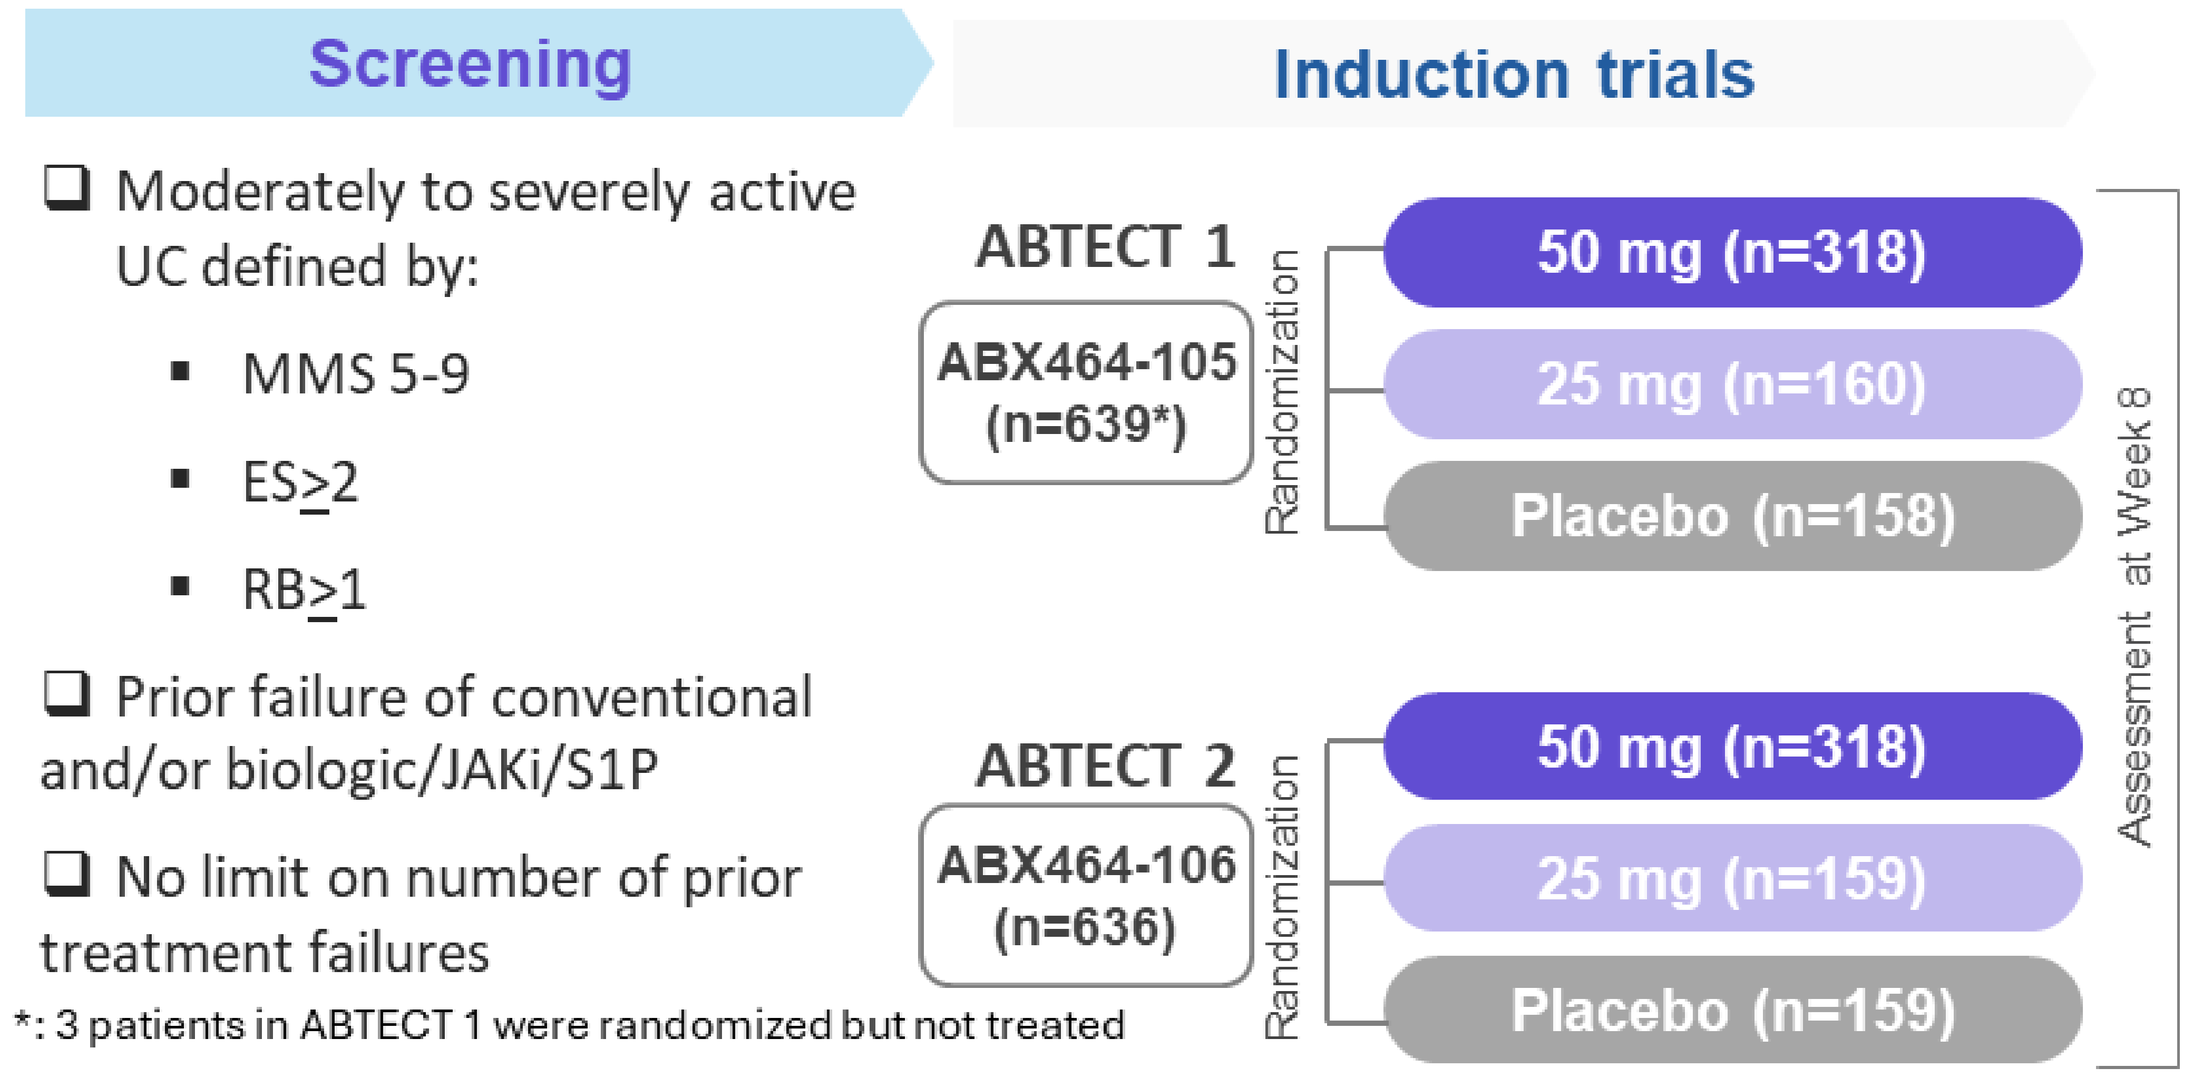
Screening
Induction trials
❑ Moderately to severely active UC defined by:
▪ MMS 5-9
▪ ES≥2
▪ RB≥1
❑ Prior failure of conventional and/or biologic/JAKi/S1P
❑ No limit on number of prior treatment failures
ABTECT 1
ABX464-105 (n=639*)
Randomization
50 mg (n=318)
25 mg (n=160)
Placebo (n=158)
ABTECT 2
ABX464-106 (n=636)
Randomization
50 mg (n=318)
25 mg (n=159)
Placebo (n=159)
Assessment at Week 8
*: 3 patients in ABTECT 1 were randomized but not treated

Methods

- Treatment emergent adverse events (TEAEs), serious TEAEs, discontinuation rates and primary reasons were assessed in the pooled population from ABTECT-1 [NCT05507203] and ABTECT-2 [NCT05507216] among pts who received Obe 50 mg QD (Obe-50), Obe 25 mg QD (Obe-25), or placebo (PBO) for 8 weeks, randomized to 2:1:1.

Results

- 1272 pts were randomized and treated in ABTECT induction trials. Baseline demographics and disease characteristics were comparable across Obe-50, Obe-25 and PBO groups with median exposures of 60, 61, and 61 days, respectively (Table 1).
- Proportions of pts who reported at least one TEAE were 60.2%, 48.9%, and 50.8% for Obe-50, Obe-25, and PBO, respectively (Table 1).
- The most frequent TEAEs included headache, nausea, abdominal pain, and lipase increased.
- TEAEs leading to study discontinuation occurred at similar rates across treatment groups (Obe-50: 4.7%, Obe-25: 1.9%, PBO: 4.1%) (Table 1).
- No signal for serious, severe, or opportunistic infections or malignancies was observed (Table 1).
- The overall rates of serious TEAEs for pts were comparable across groups (Obe-50: 3.1%, Obe-25: 2.2%, PBO: 3.2%) (Tables 1 & 2).
- No clustering of serious TEAEs was observed; only worsening of UC and pneumonia were reported more than once in any individual treatment arm (Table 2).

Table 2: Serious TEAEs in ABTECT Induction Trials

	Obe-50 (n=636)	Obe-25 (n=319)	PBO (n=317)
Serious TEAE, n (%)	**20 (3.1)**	**7 (2.2)**	**10 (3.2)**
Colitis ulcerative	5 (0.8)	2 (0.6)	5 (1.6)
Gastritis	1 (0.2)	0	0
Pancreatitis	1 (0.2)	0	0
Pancreatitis acute	1 (0.2)	1 (0.3)	1 (0.3)
Vomiting	1 (0.2)	0	0
Anal inflammation	0	0	1 (0.3)
Pneumonia	2 (0.3)	0	0
Anal abscess	1 (0.2)	0	0
Appendicitis	0	1 (0.3)	0
Clavicle fracture	1 (0.2)	0	0
Procedural dizziness	1 (0.2)	0	0
Anaemia	1 (0.2)	0	0
Blood loss anaemia	0	0	1 (0.3)
Myocardial infarction	1 (0.2)	0	0
Acute coronary syndrome	0	0	1 (0.3)
Type 2 diabetes mellitus	1 (0.2)	0	0
Prostate cancer stage I	1 (0.2)	0	0
Migraine	1 (0.2)	0	0
Calculus bladder	1 (0.2)	0	0
Pyrexia	0	0	1 (0.3)
Lipase increased	0	1 (0.3)	0
Pleurisy	0	1 (0.3)	0
Vasculitis	0	0	1 (0.3)

Table 1: Integrated Summary of Adverse Events of Obe in Phase 3 ABTECT Induction Trials: Safety Set

	Obe-50 (n=636)	Obe-25 (n=319)	PBO (n=317)
Duration of exposure, days, mean (SD)	58.8 (12.6)	59.5 (11.1)	58.7 (11.1)
Median	60.0	61.0	61.0
[Min, Max]	[1, 106]	[4, 96]	[5, 102]
Any TEAEs, n (%)	383 (60.2)	156 (48.9)	161 (50.8)
TEAE related to study drug, n (%)	226 (35.5)	86 (27.0)	55 (17.4)
TEAE leading to study discontinuation, n (%)	30 (4.7)	6 (1.9)	13 (4.1)
Related TEAE leading to study discontinuation, n (%)	20 (3.1)	4 (1.3)	6 (1.9)
Serious TEAE, n (%)	20 (3.1)	7 (2.2)	10 (3.2)
Related serious TEAE, n (%)	3 (0.5)	2 (0.6)	2 (0.6)
TEAE leading to death, n (%)	0	0	0
Malignancies	1 (0.2)[a]	0	0
Serious/severe/opportunistic infections	4 (0.6)[§]	1 (0.3)[¥]	1 (0.3)[‡]
TEAEs occurring in patients (≥3% in Obe groups and greater than PBO), n (%)			
Headache	148 (23.3)	48 (15.0)	18 (5.7)
Nausea	46 (7.2)	16 (5.0)	4 (1.3)
Abdominal pain	24 (3.8)	2 (0.6)	2 (0.6)
Lipase increased[b]	27 (4.2)	9 (2.8)	7 (2.2)

a: Prostate cancer stage I. b: To date, no safety signals have been observed related to either elevations in lipase or more specifically to pancreatitis.
§: Pneumonia (2), anal abscess, COVID-19; ¥: Appendicitis; ‡: Bronchopulmonary aspergillosis.

- Headache TEAEs were mild, transient, short in duration (median: 2-3 days) and rarely led to discontinuation (0-1.1%) (Tables 3 & 4).

Table 3: Headaches in ABTECT Induction Trials

	Obe-50 (n=636)	Obe-25 (n=319)	PBO (n=317)
Time to onset of first TE headache per patient, days, median	1.0	1.0	7.0
Duration of headache for all TE headaches, days, median	2.0	3.0	2.0

- Rates of pts who discontinued the study occurred at similar rates across treatment groups (Table 4).

Table 4: Discontinuations in ABTECT Induction Trials

	Obe-50 (n=636)	Obe-25 (n=319)	PBO (n=317)
Patients who discontinued the study, n (%)[a]	54 (8.5)	23 (7.2)	31 (9.7)
Reason for study discontinuation, n (%)[b]			
Adverse event	31 (57.4)	8 (34.8)	14 (45.2)
Withdrawal by subject	13 (24.1)	10 (43.5)	10 (32.3)
Lack of efficacy	2 (3.7)	2 (8.7)	1 (3.2)
Lost to follow-up	4 (7.4)	1 (4.3)	1 (3.2)
Noncompliance with study drug	2 (3.7)	0	0
Protocol deviation	1 (1.9)	1 (4.3)	3 (9.7)
Physician decision	1 (1.9)	0	1 (3.2)
Never dosed	0	1 (4.3)	1 (3.2)
TEAEs (>1 in Obe groups) leading to study drug discontinuation			
Ulcerative colitis	7 (1.1)	2 (0.6)	7 (2.2)
Headache	7 (1.1)	1 (0.3)	0
Vomiting	4 (0.6)	1 (0.3)	0
Abdominal pain upper	3 (0.5)	0	0
Nausea	3 (0.5)	1 (0.3)	0
Pancreatitis acute	2 (0.3)	1 (0.3)	1 (0.3)

a: Percentages are calculated relative to the number of randomized patients (ie, n=636, 320, 319 for Obe-50, Obe-25, PBO, respectively).
b: Percentages are calculated relative to the number of patients who discontinued the study.

Conclusions

This integrated safety analysis of the two completed Phase 3 ABTECT induction trials in moderately to severely active UC showed a favorable safety and tolerability profile of obefazimod as observed in Phase 2 trials.

References 1: Vermeire S et al. *J Crohns Colitis*. 17: 1689-97, 2023 - 2: Vermeire S et al. *Gastroenterology*. 160: 2595-98, 2021 - 3: Vermeire S et al. *The Lancet Gastroenterology & Hepatology*. 7: 1024-35, 2022

Disclosures : US *(consultant or speaker's fees) AbbVie, Abivax, Amgen, Galapagos, Janssen, Eli Lilly, Janssen; (Grant) Abbvie, Abivax, Boehringer Ingelheim, BMS, Celgene, Eli Lilly, Gilead Sciences, Galapagos, Janssen, Pfizer, Roche, and Takeda Pharmaceuticals;* SD *(consultant or speaker's fees): AbbVie, Ferring, Hospira, Johnson & Johnson, Merck, MSD, Takeda, Mundipharma, Pfizer Inc, Tigenix, UCB Pharma, Vifor, Biogen, Celgene, Allergan, Celltrion, Sandoz, Boehringer Ingelheim;* LPB *(consultant or speaker's fees)* Abbvie, Abivax, Adacyte, Alimentiv, Amgen, Applied Molecular Transport, Arena, Banook, Biogen, BMS, Celltrion, Connect Biopharm, Cytoki Pharma, Enthera, Ferring, Fresenius Kabi, Galapagos, Genentech, Gilead, Gossamer Bio, GSK, IAC Image Analysis, Index Pharmaceuticals, Inotrem, Janssen, Lilly, Medac, Mopac, Morphic, MSD, Nordic Pharma, Novartis, Oncodesign Precision Medicine, ONO Pharma, OSE Immunotherapeuthics, Pandion Therapeuthics, Par' Immune, Pfizer, Prometheus, Protagonist, Roche, Samsung, Sandoz, Sanofi, Satisfay, Takeda, Telavant, Theravance, Thermo Fischer, Tigenix, Tillots, Viatris, Vectivbio, Ventyx, Ysopia (Grant) Celltrion, Fresenius Kabi, Medac, MSD, Takeda; MD *(consultant/speaker's fees)* Abbvie, Abivax, Arena Pharmaceuticals, Astra Zeneca, Boehringer Ingelheim International GmbH, Bristol-Meyer Squibb, Eli Lilly and Company, F. Hoffmann-La Roche Ltd, Genentech Inc, Gilead, Janssen Pharmaceuticals, Merck, Pfizer Inc, Prometheus Biosciences, Takeda Pharmaceuticals; RA *(consultant or speaker's fees) AbbVie, Abivax, AstraZeneca, BMS, Celltrion Healthcare, Galapagos, J&J, Lilly, MSD, Pfizer, and Takeda Pharma;* AA *(consultant or speaker's fees) Abivax, Alfa Sigma, Astra Zeneca, Biogen, Boehringer Ingelheim, Bristol-Myers Squibb, Celltrion, Eli-Lilly, Enthera, Ferring, Galapagos, Gilead, Giuliani, Janssen, Lionhealth, MSD, Nestlé, Pfizer, Protagonist Therapeutics, Roche, Samsung Bioepis, Sanofi, Sandoz, Takeda, Teva Pharmaceuticals, Tillots Pharma, AG Pharma, Novartis; (Grant) Biogen, MSD, Takeda, Pfizer;* DL *(consultant or speaker's fees) Abbvie, Alfasigma, Amgen, Biocon, Celltrion, Ferring, Fresenius-Kabi, Johnson & Johnson, Lilly, Medac, MSD, Pfizer, Sandoz, Takeda;* PSD *(consultant or speaker's fees) Abbvie, Abivax, Adiso, Alimentiv, Bristol Meyer Squibb, Celltrion, Genentech, Geneoscopy, Janssen, Pfizer, Takeda;* DR *(consultant or speaker's fees)* Abbvie, Abivax SA, Altrubio, Athos Therapeutics, Inc, Bristol-Myers Squibb, Celltrion, Connect BioPharma, Eli Lilly, Genentech, Iterative Health, Janssen Pharmaceuticals, J&J, Merck & Co., Odyssey Therapeutics, Pfizer, Sanofi, Spyre, Takeda Pharmaceuticals, Vedanta Biosciences, Ventyx (Grant) Takeda; BES *(consultant/speaker's fees) AbbVie, Abivax, Adiso Therapeutics, AgomAb, Alimentiv, Amgen, Arena Pharmaceuticals, Artizan Biosciences, Artugen Therapeutics, AstraZeneca, Bacainn Therapeutics, Biora Therapeutics, Boehringer Ingelheim, Boston Pharmaceuticals, Bristol Myers Squibb, Calibr, Celltrion, ClostraBio, Connect Biopharm, Cytoki Pharma, Eli Lilly and Company, Enthera, Evommune, Ferring, Fresenius Kabi, Galapagos, Gilead Sciences, Genentech, Glaxo SmithKline, Gossamer Bio, HMP Acquisition, Imhotex, Immunic, InDex Pharmaceuticals, Innovation Pharmaceuticals, Inotrem, Ironwood Pharmaceuticals, Janssen, Johnson & Johnson, Kaleido, Kalyope, Merck, MiroBio, Morphic Therapeutic, MRM Health, OSE Immunotherapeutics, Pfizer, Progenity, Prometheus Biosciences, Prometheus Laboratories, Protagonist Therapeutics, Q32 Bio, RedHill Biopharma, Sun Pharma Global, Surrozen, Synlogic Operating Company, Takeda, Target RWE, Theravance Biopharma R&D, TLL Pharmaceutical, USWM Enterprises, Ventyx Biosciences, Viela Bio, and stock options from Ventyx Biosciences.*

Copyright © 2026 Abivax

7-11 Blvd Haussmann- 75009 Paris, France
E-mail: info@abivax.com - www.abivax.com